Factors Influencing Best Practices in Treating Ulcerative Colitis: Results from a Predictive Modeling Analysis of Educational Outcomes Data

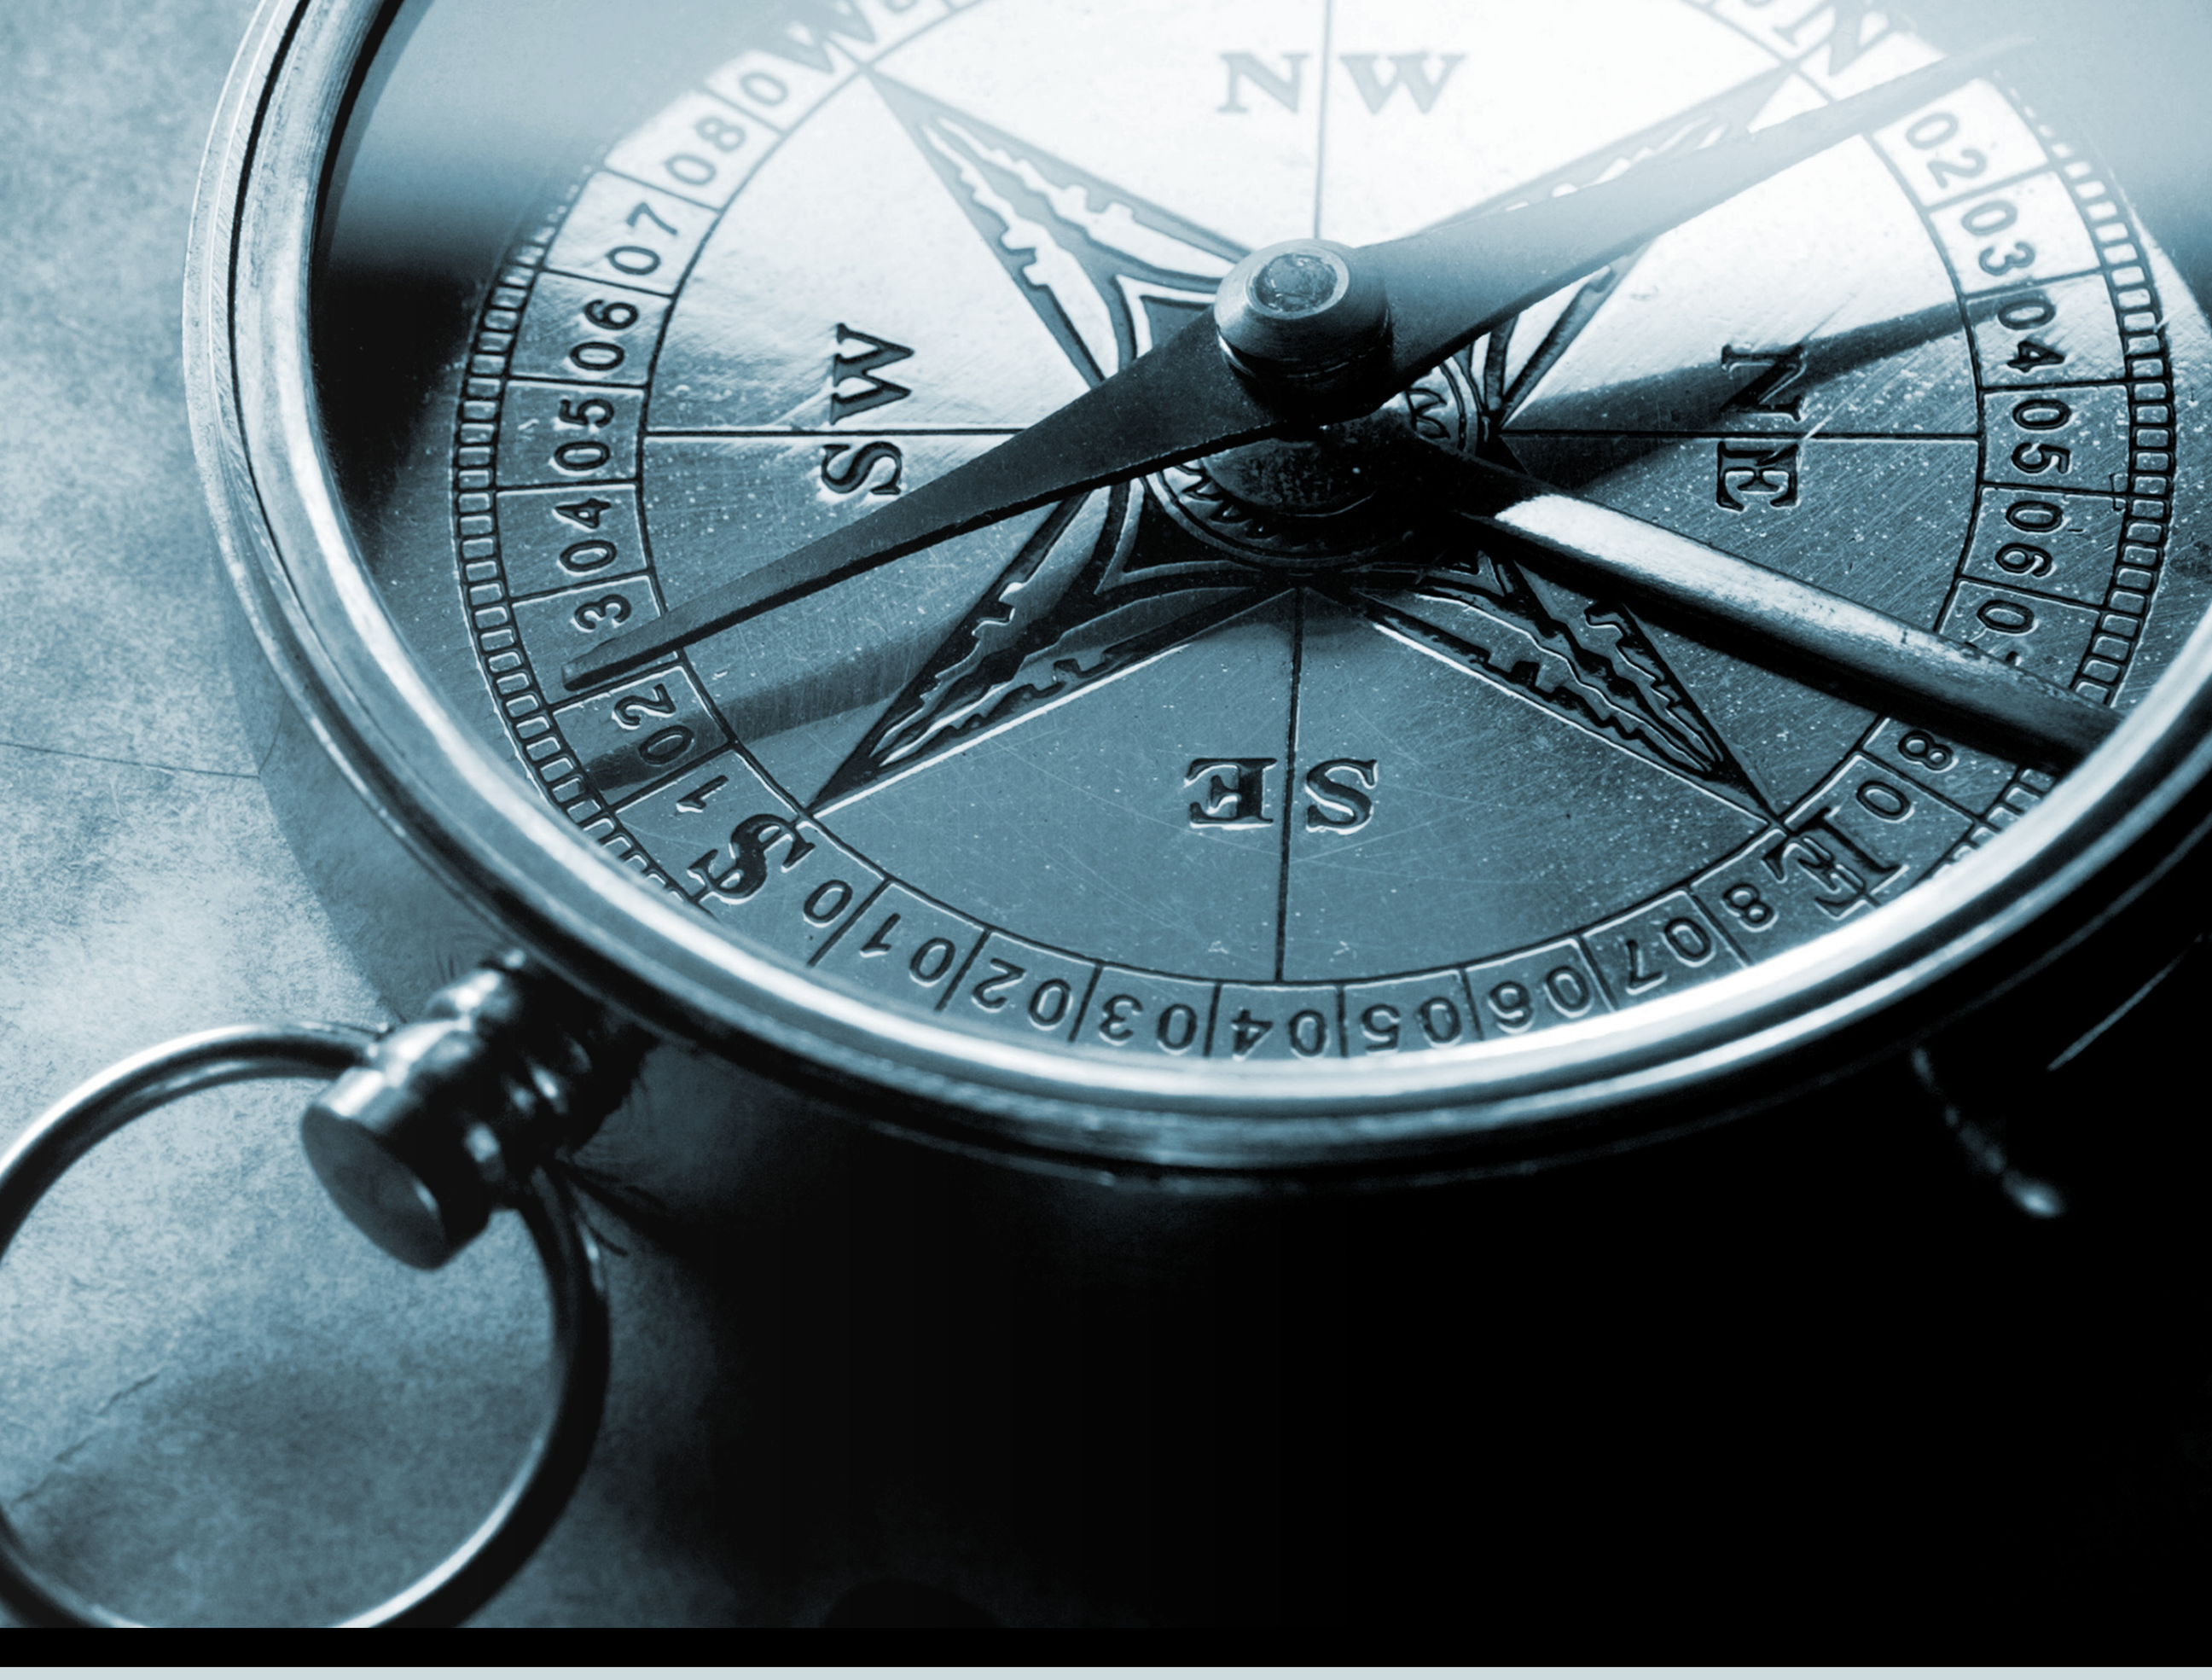

Jamie Reiter, PhD[1] • Jan Perez[1] • Sharon A. Tordoff, BS[1] • Whitney E. Faler, MPA[1]

[1]CME Outfitters, LLC

INTRODUCTION

An essential component of improving patient outcomes through medical education is ensuring healthcare providers (HCPs) perform according to best practices. Traditional statistical comparisons of pre- versus post-activity performance are important for demonstrating performance improvement. However, they do not provide information regarding the factors that influence practice behaviors; if an activity was successful in changing HCP behavior, do we know why? Conversely, if an activity was not successful, what may be the barrier or reason preventing improvements? Understanding what influences these improvements or lack thereof can help us develop future activities that continue what was successful or make necessary changes in our processes. Both scenarios can result in maximally effective educational activities which will ultimately improve patient outcomes. PredictCME is CME Outfitters' exclusive method for applying a predictive modeling technique, known as CHAID (chi-square automatic interaction detection),[1] to our educational activities. This presentation provides results from a PredictCME analysis of behavior data from an educational activity on ulcerative colitis (UC).

BACKGROUND

A Brief Primer on Prediction

Predictive modeling is frequently used in various research settings, but it is rarely used in medical education. The most common prediction method is regression, which is a method for predicting values of a "response" or "criterion" variable from the values of one or more "predictor" variables. Linear regression is used when predictor and response variables are continuous (e.g., age, weight) and logistic regression is used for response variables that are categorical (e.g., correct/incorrect).

Both linear and logistic regression are commonly used in statistics, and both have their strengths. However, among some of their limitations are flexibility and interpretability.

CHAID/PredictCME

CHAID is a form of predictive modeling, often used in data mining, which can be used for both continuous and categorical data. Output is in the form of a classification (or decision) tree, which provides a visual representation of the interplay between predictor and response variables, as well as how the variable categories are broken down. PredictCME is CME Outfitters' exclusive method for applying CHAID to our educational activities, so that we can design future activities with a scientific basis for what impacts performance. Results from PredictCME will help guide needs assessments and ensure the appropriate topics, formats, questions, and audiences are targeted.

In addition to predicting factors that influence performance, PredictCME can be used for determining which variables most impact knowledge, confidence, competence, or other endpoints. It should also be noted that predictive modeling is used for data from a single time point rather than comparing data from two or more time points. Table 1 outlines the possible outcomes survey time points and corresponding response variablesto consider when performing predictive modeling.

Time Point	Possible Response Variables
Pre-survey(participants)	Existing knowledge, confidence, competence, behavior
Post-survey (participants)	Immediate post-activity knowledge, confidence, competence, planned behavior
Follow-up survey (participants)	Longer-term knowledge, confidence, competence, behavior
Follow-up survey (controls)	Similar to pre-survey
Change scores (matched participants; pre/post, pre/ follow-up, post/follow-up)	Changes in knowledge, confidence, competence, behavior

Table 1. Outcomes Survey Time Points and Corresponding Response Variables for Predictive Modeling

METHODS

Educational outcomes data were obtained from an educational activity on UC, which consisted of a live and streamed symposium at Digestive Diseases Week 2017, and integrated interactive infographics to facilitate content delivery and translation. Surveys assessing knowledge, confidence, and behavior were administered before and immediately following the activity, to establish baseline as well as any improvements as a result of the activity. A separate evaluation survey was also administered immediately following the activity, which provided demographics and other variables used in the model.

An analysis using PredictCME was conducted on data from the pre-activity survey, which included two behavior questions related to applying unique risk/benefit profiles of biologic therapies to individualizing treatment and utilizing data from real-world studies of biologic therapy to initiate early treatment. Data from these two questions were converted to a single behavior score (as described below) and used as the response variable in the analysis. Demographics, knowledge, confidence, and evaluation data were entered as predictors.

Data from the following two behavior questions were combined into an aggregate "behavior" score, as follows. For each question, if participants selected "76% to 100% of the time," that was scored as a "1," and 0 otherwise. These 1's and 0's were added across the two behavior questions, so possible scores for the aggregate behavior score were 0, 1, or 2. This aggregate score was used as the response variable in the PredictCME analysis.

Behavior questions used for the aggregate behavior score:

How often do you apply the unique risk/benefit profiles of biologic therapies when making treatment decisions based on individual prognosis and severity of disease?

a. 0% of the time
b. 1% -25% of the time
c. 26% -50% of the time
d. 51% -75% of the time
e. 76% -100% of the time

How often do you utilize data from real-world studies on the use and effectiveness of biologic therapy for UC to initiate early, effective treatment for your patients with UC?

a. 0% of the time
b. 1% -25% of the time
c. 26% -50% of the time
d. 51% -75% of the time
e. 76% -100% of the time

Several predictor variables* were entered into the model:

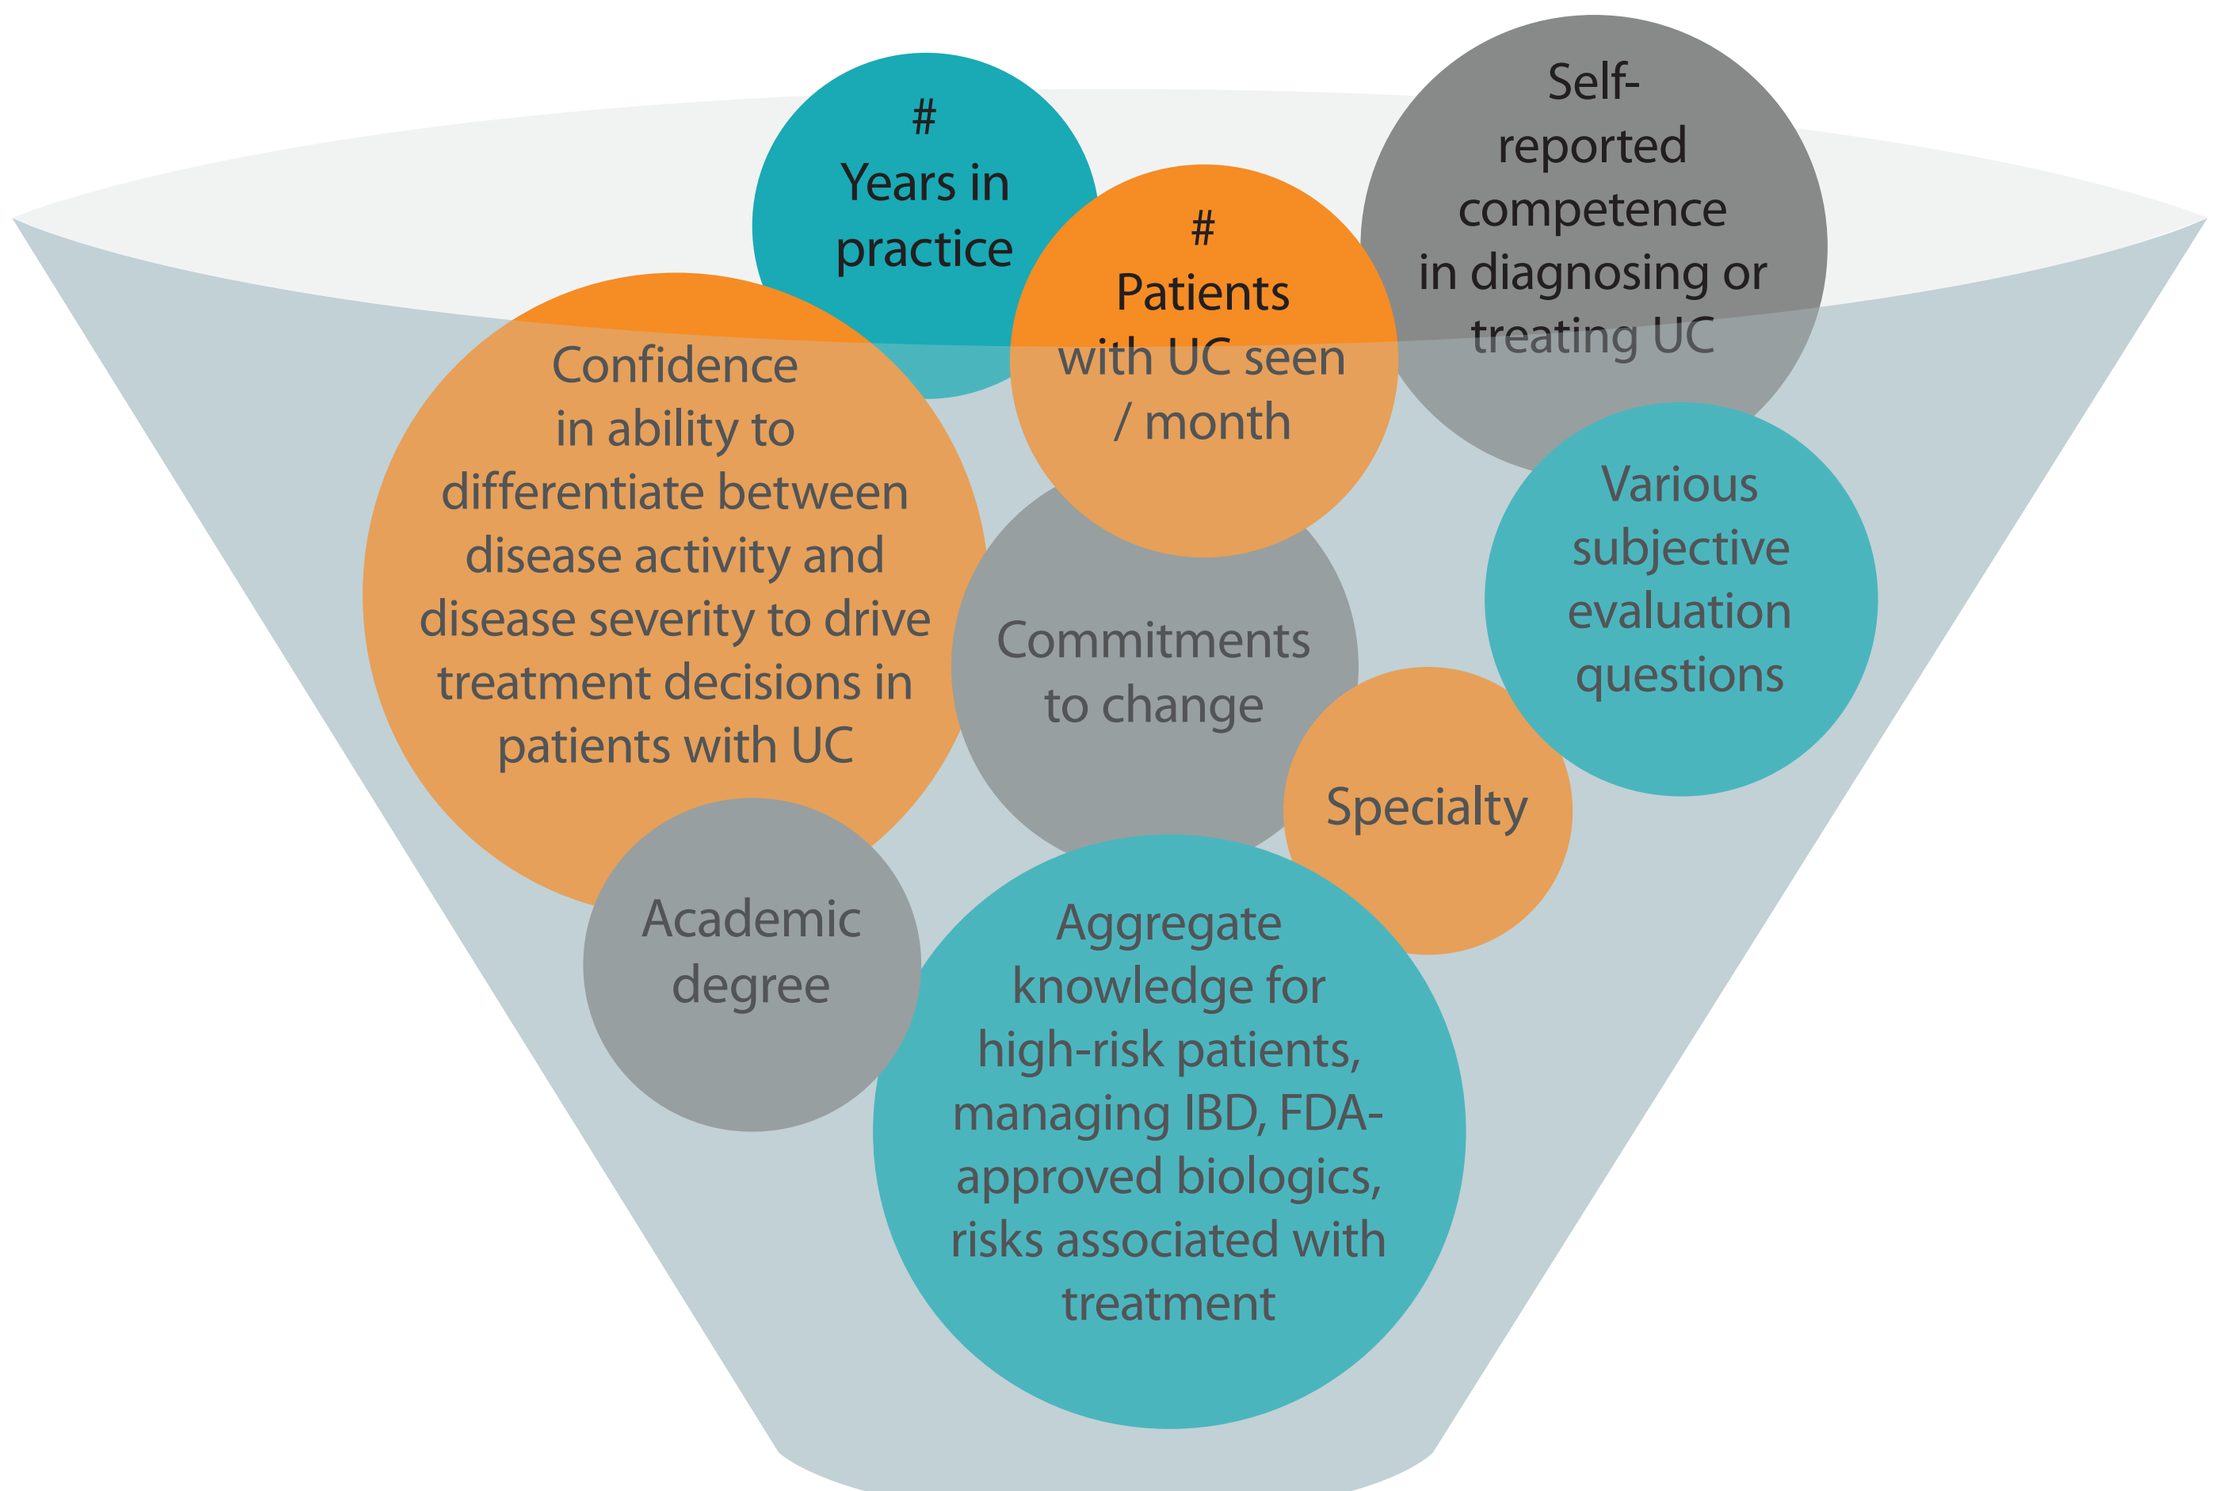

Years in practice
Patients with UC seen / month
Self-reported competence in diagnosing or treating UC
Confidence in ability to differentiate between disease activity and disease severity to drive treatment decisions in patients with UC
Commitments to change
Various subjective evaluation questions
Specialty
Academic degree
Aggregate knowledge for high-risk patients, managing IBD, FDA-approved biologics, risks associated with treatment

* Predictor variables were selected based on expert assessments on which would most likely influence behavior and which variables would be of most interest. As CME Outfitters is the first provider to utilize this technique in medical education, there are currently no established algorithms or references guiding variable selection. We are in the process of developing such guidelines. In addition, although not available for this activity, data from questions related to practice barriers would be an important component of predictive models, which we will be incorporating in future PredictCME analyses.

RESULTS

Over 400 clinicians participated in the activity, with 119 participating in the pre-survey used for the analysis. Figure 1 shows the breakdown of specialty, academic degrees and years in practice of the participants.

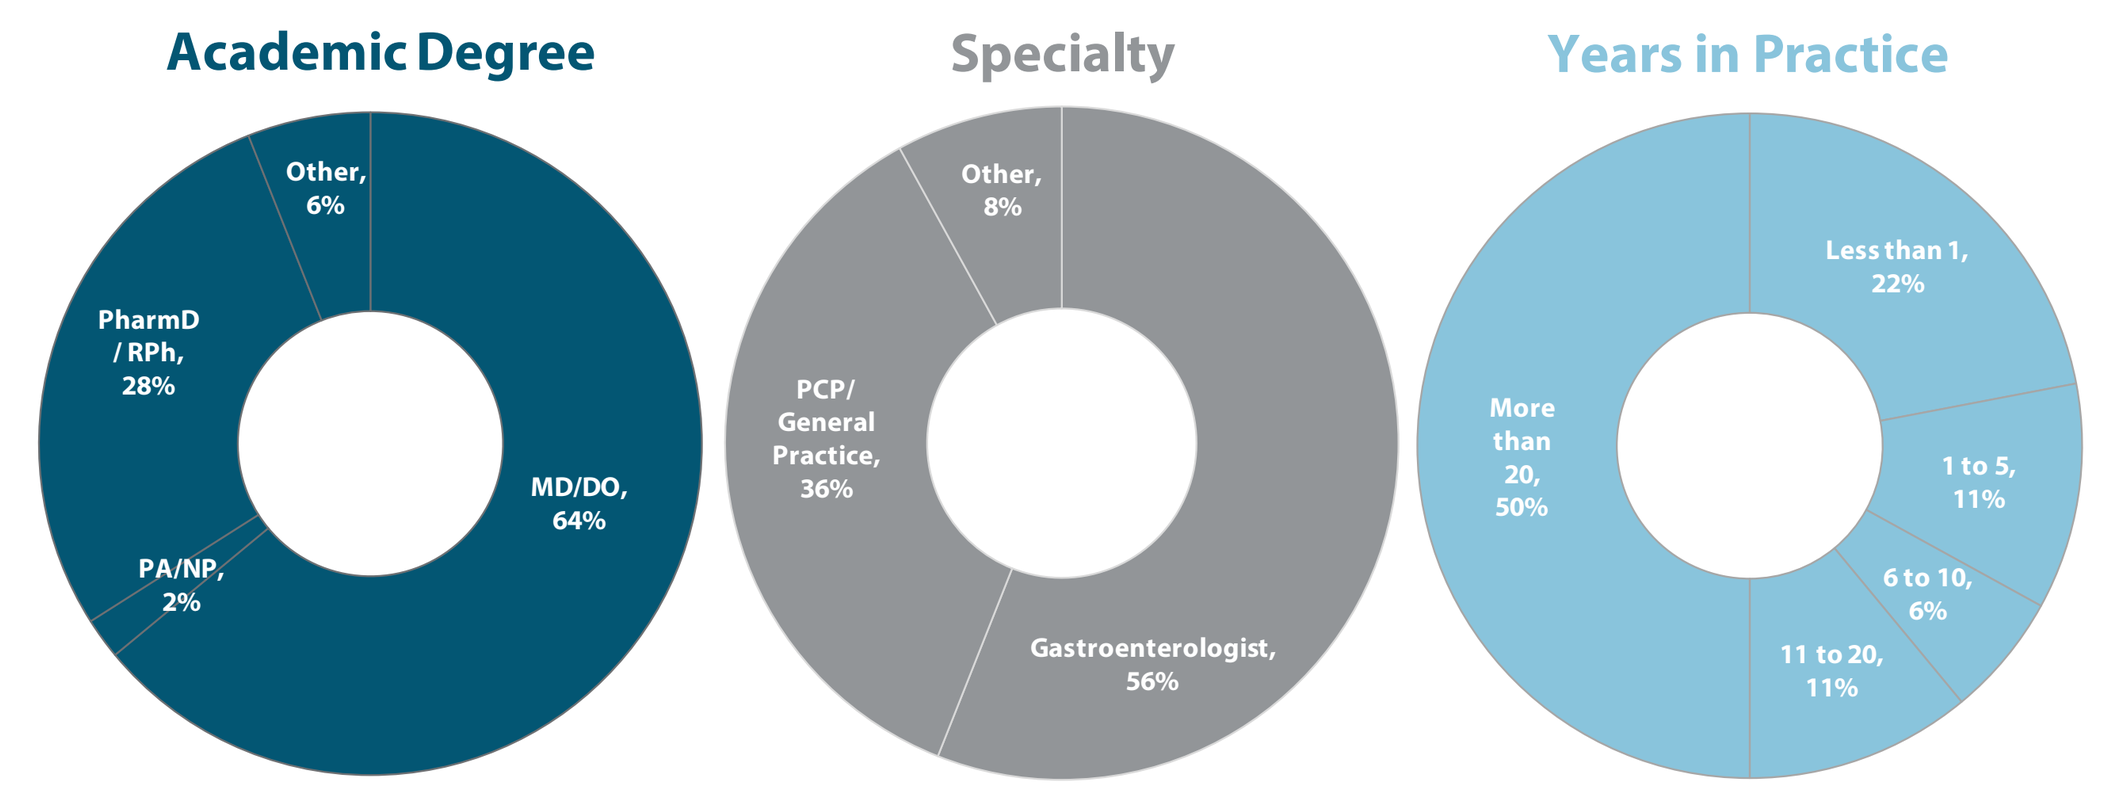

Academic Degree
Other, 6%
PharmD / RPh, 28%
MD/DO, 64%
PA/NP, 2%
Specialty
Other, 8%
PCP/ General Practice, 36%
Gastroenterologist, 56%
Years in Practice
Less than 1, 22%
1 to 5, 11%
6 to 10, 6%
11 to 20, 11%
More than 20, 50%

Figure 1. Demographic Distributions for Participants in an Educational Activity on Ulcerative Colitis.

Figure 2 shows the PredictCME output for the aggregate behavior score in tree format. All graphs in the output reflect percentages of participants who performed 0, 1, or 2 of the two behaviors at least 76% of the time. Results are best interpreted by comparing the percentages between the top and bottom right-most graphs.

Interpretation of the output would be as follows:

1. Overall, 33% of participants at the pre-survey performed at least one behavior at least 76% of the time (left-most graph).

2. The primary, or strongest, predictor of performing these behaviors was confidence ($x^2(2) = 7.04$, $p < .05$). The right two graphs reflect how the data were broken down, based on how the model maximized the difference. A greater percentage of participants who were "Confident" or "Very Confident" performed at least one behavior (41%) compared to those who were "Somewhat" or "Not at all Confident" (21%).

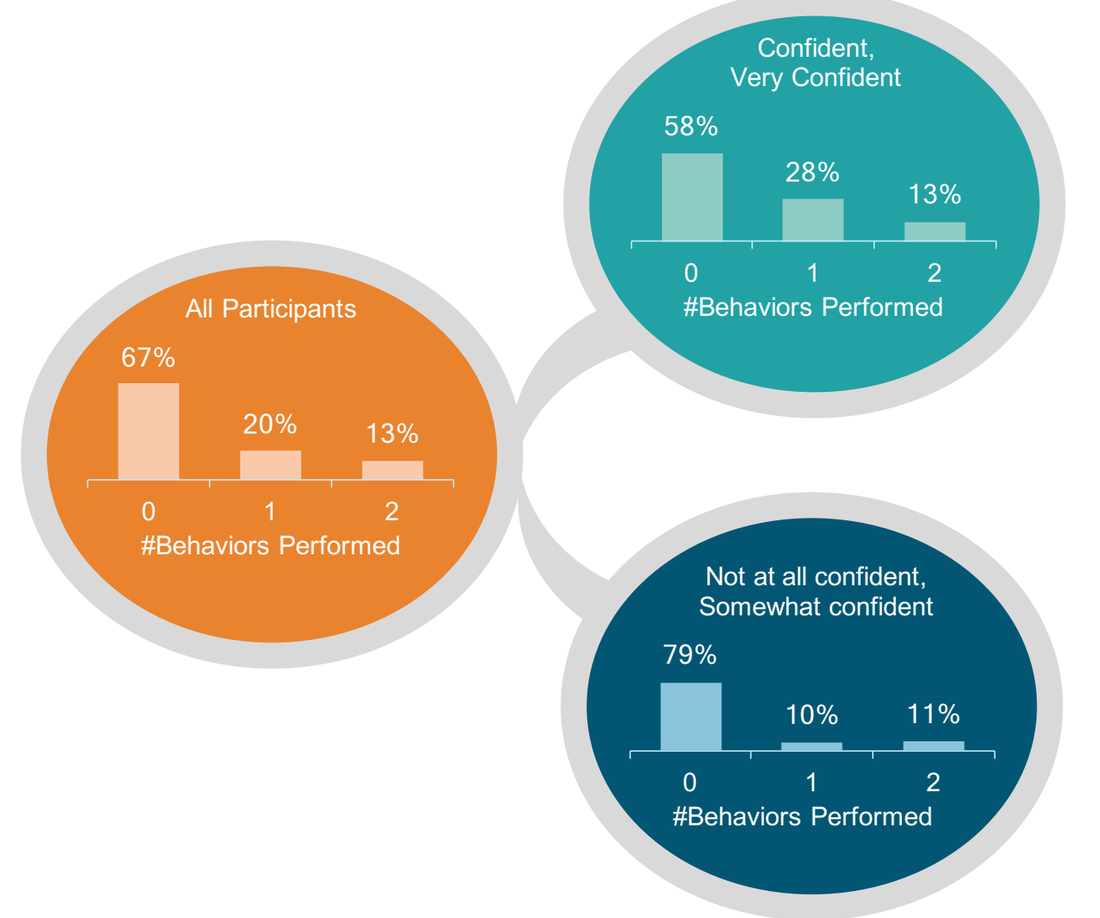

All Participants
67%
20%
13%
0
1
2
#Behaviors Performed
Confident, Very Confident
58%
28%
13%
0
1
2
#Behaviors Performed
Not at all confident, Somewhat confident
79%
10%
11%
0
1
2
#Behaviors Performed

Figure 2. PredictCME Output for Pre-Survey Aggregate Behavior Score

To further explore the data, we conducted separate PredictCME analyses on each behavior question. For the behavior question regarding applying risk/benefit profiles of biologic therapies when making treatment decisions, confidence was again the strongest predictor ($x^2(1) = 5.69$, $p < .05$), with 35% of those who were confident or very confident performing the behavior versus 15% of those who were somewhat or not at all confident (Figure 3). For the behavior question regarding utilizing data from real-world studies of biologic therapy for UC, aggregate performance across the four knowledge questions was the strongest predictor. Specifically, participants achieving at least 70% correct were more likely to perform the behavior compared to those who achieved less than 70% correct (53% versus 15%, respectively, $x^2(1) = 11.98$, $p < .05$) (Figure 4). Another observation for this behavior question is that participants who achieved at least 70% correct were almost equally likely to perform the behavior (53%) or not perform the behavior (47%).

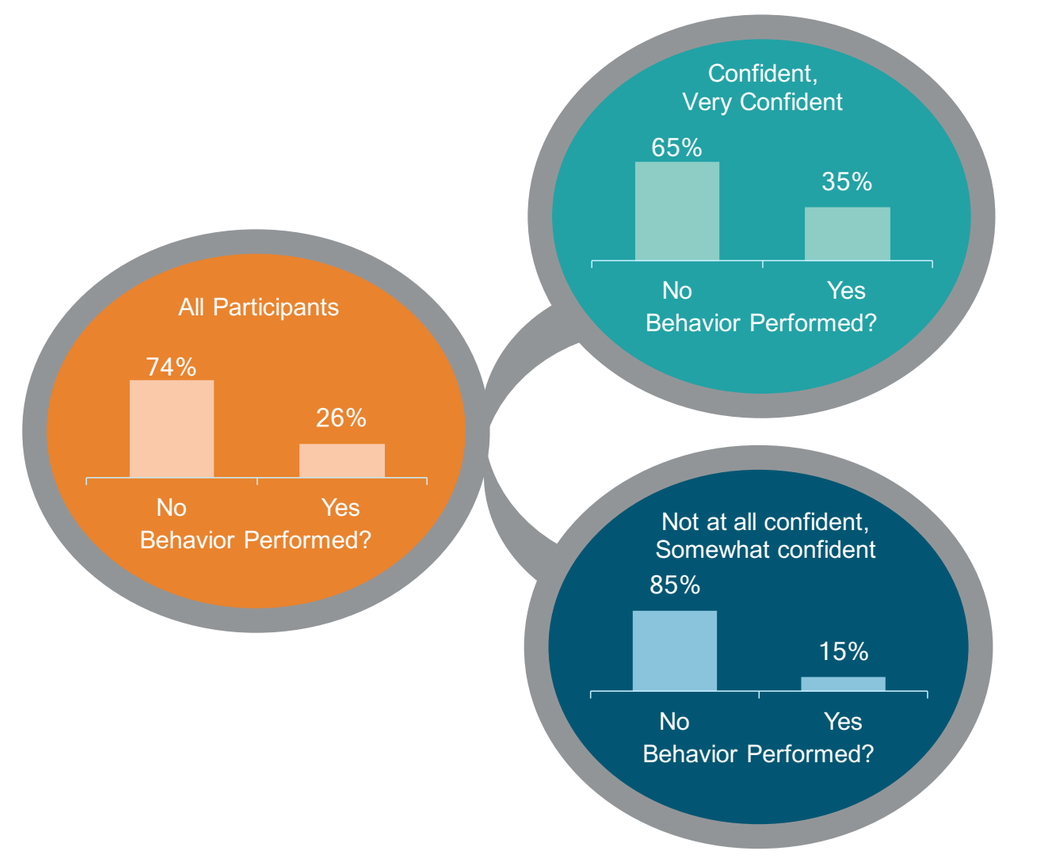

All Participants
74%
26%
No
Yes
Behavior Performed?
Confident, Very Confident
65%
35%
No
Yes
Behavior Performed?
Not at all confident, Somewhat confident
85%
15%
No
Yes
Behavior Performed?

Figure 3. PredictCME Output for Pre-Survey Behavior Question Regarding Risk/Benefit Profiles

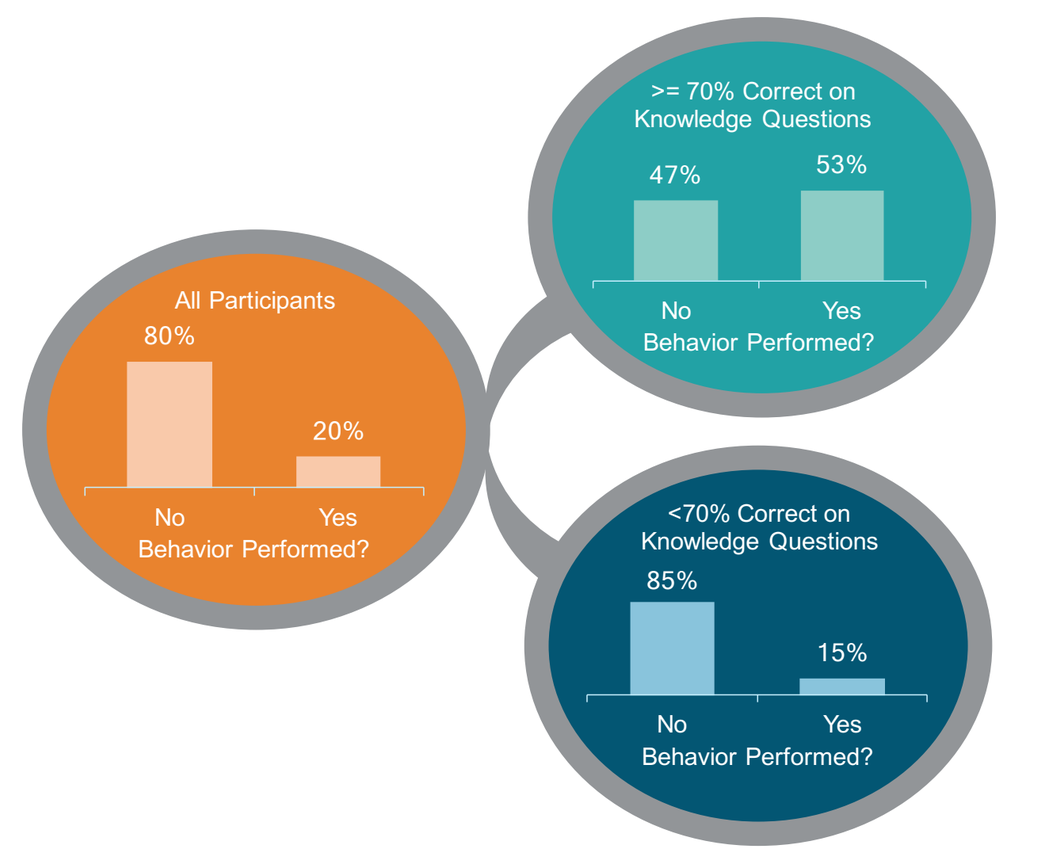

All Participants
80%
20%
No
Yes
Behavior Performed?
>= 70% Correct on Knowledge Questions
47%
53%
No
Yes
Behavior Performed?
<70% Correct on Knowledge Questions
85%
15%
No
Yes
Behavior Performed?

Figure 4. PredictCME Output for Pre-Survey Behavior Question Regarding Utilizing Data from Real-World Studies

RESULTS (cont.)

Discussion

- Results from the PredictCME analysis were not surprising, as early behavior studies found confidence to influence behavior.[2] In addition, previous analyses using PredictCME also found confidence to be the strongest predictor of behavior in medical education activities.[3-6]
- With regard to the behavior question related to utilizing data from real-world studies of biologic therapy for UC, the finding that participants who were more knowledgeable were almost equally likely to perform the behavior than not, indicates that knowledge exerts its influence on behavior primarily when there is a lack of knowledge.
- What these results also highlight is that the motivation for performing certain practice behaviors can differ depending on the specific behavior. In this case, confidence related to treatment decisions predicted behavior related to making treatment decisions, which makes intuitive sense. It is also somewhat intuitive that knowledge would predict a behavior related to translating clinical data to practice.
- Given that confidence was the strongest predictor for the aggregate behavior score, the assumption would be that confidence had a stronger influence overall.

The findings from this study are currently being integrated into our planning for future PredictCME analyses as well as educational activities. For future activities, we are evaluating ways to improve aspects of HCP confidence and knowledge that are most likely to influence best practice behaviors.

CONCLUSIONS

These findings from the PredictCME analysis demonstrate the utility in using predictive modeling to better understand the influences of practice behavior. We prefer PredictCME to regression, as the procedure is more flexible, and the output is more intuitive and informative. It is our hope that other medical education providers will utilize predictive modeling, in its various forms, to help determine the factors that help or hinder the success of their educational activities, which in turn will help maximize the impact of future activities, and ultimately patient outcomes.

REFERENCES

1. Kass GV. An exploratory technique for investigating large quantities of categorical data. Applied Statistics.1980;29(2):119-127.
2. Bandura A. Self-efficacy: toward a unifying theory of behavioral change. Psychol Rev.1977;84:191-215.
3. Reiter J, Perez J, Tordoff S, Faler W. Novel Approaches to Predictive Modeling for Customizing Educational Activities. Poster presented at: SACME Annual Meeting; May 16-20, 2017; Scottsdale, AZ.
4. Reiter J, Perez J, Tordoff S, FalerW. Factors Influencing Best Practices in Treating Multiple Sclerosis: Results from a Predictive Modeling Analysis. Poster accepted for presentation at: MSParis2017: 7th Joint ECTRIMS-ACTRIMS Meeting; October 25-28, 2017; Paris, France.
5. Reiter J, Perez J, Tordoff S, Faler W. Factors Influencing Best Practices in Treating IBD: Results from a Predictive Modeling Analysis of Educational Outcomes Data. Poster accepted for presentation at: 2017 Advances in Inflammatory Bowel Diseases (AIBD) Conference;November 9-11,2017; Orlando, FL.
6. Reiter J, Perez J, Tordoff S, Faler W. Factors Influencing Shared Decision Making in Developing a Seizure Action Plan: Results from a Predictive Modeling Analysis of Educational Outcomes Data. Poster accepted for presentation at: American Epilepsy Society (AES) Annual Meeting; December 1-5, 2017; Washington, DC

FUNDING

The educational activity described in this presentation was supported by an educational grant from Takeda Pharmaceuticals U.S.A., Inc.

CME Outfitters CONTINUING MEDICAL EDUCATION

Scan the QR code on the right to view a pdf of this poster online.

© CME Outfitters, LLC, 2017. All rights reserved.