Factors Influencing Shared Decision Making in Developing a Seizure Action Plan: Results from a Predictive Modeling Analysis of Educational Outcomes Data

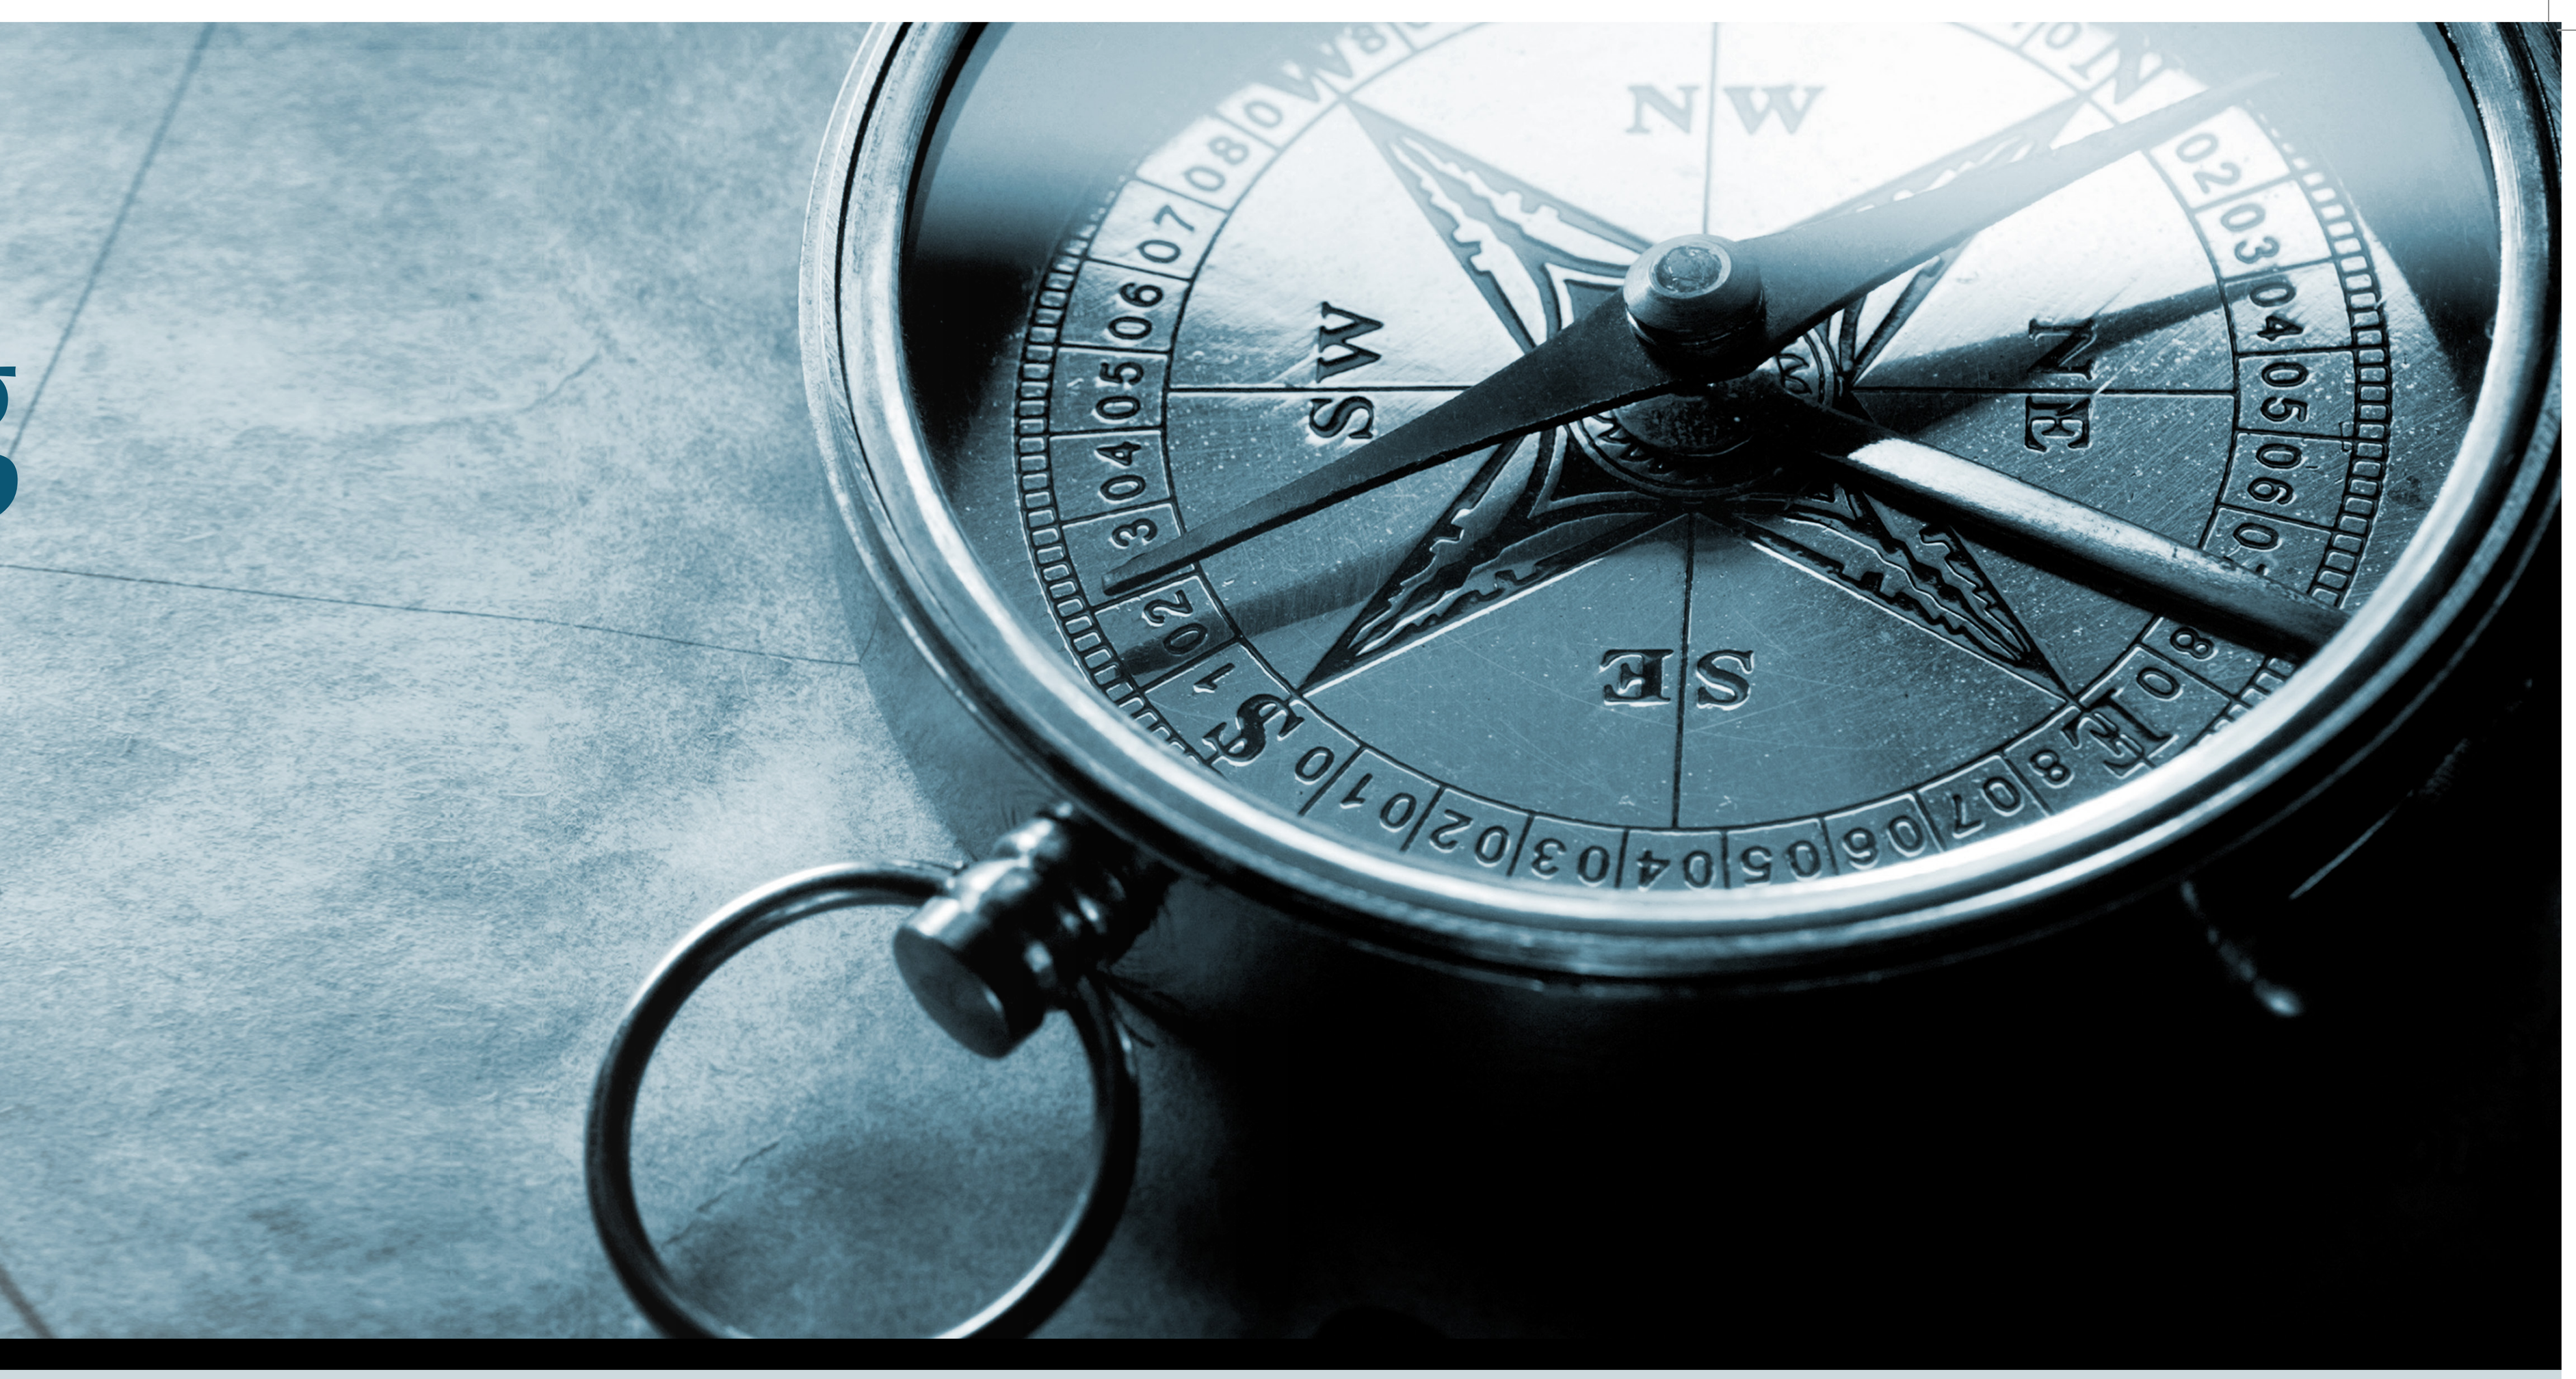

Jamie Reiter, PhD[1] • Jan Perez[1] • Sharon A. Tordoff, BS[1] • Whitney E. Faler, MPA[1]
[1]CME Outfitters, LLC

INTRODUCTION

An essential component of improving patient outcomes through medical education is ensuring healthcare providers (HCPs) perform according to best practices. Traditional statistical comparisons of pre- versus post-activity performance are important for demonstrating performance improvement. However, they do not provide information regarding the factors that influence practice behaviors; if an activity was successful in changing HCP behavior, do we know why? Conversely, if an activity was not successful, what may be the barrier or reason preventing improvements?

Understanding what influences these improvements or lack thereof can help us develop future activities that continue what was successful or make necessary changes in our processes. Both scenarios can result in maximally effective educational activities which will ultimately improve patient outcomes.

PredictCME is CME Outfitters' exclusive method for applying a predictive modeling technique, known as CHAID (chi-square automatic interaction detection),[1] to our educational activities. This presentation provides results from a PredictCME analysis of behavior data from an educational activity on developing a seizure action plan for patients with epilepsy.

BACKGROUND

Seizure Action Plans

Recognizing acute repetitive seizures (ARS) and having a Seizure Action Plan (SAP) that is developed through shared decision-making can improve quality of life for the millions of individuals impacted by epilepsy. Emerging treatments have the potential to enhance available options, thereby altering the treatment landscape reflected in SAPs. Continuing medical education (CME) has an opportunity to play a valuable role as a key stakeholder assisting HCPs to integrate shared decision-making into the development of much needed SAPs for patients with different seizure types. However, HCPs face several challenges when managing patients with ARS, and it is important to understand the barriers preventing HCPs from implementing best practices. The goal of this study was to utilize predictive modeling to determine factors influencing implementation of SAPs so that any barriers may be addressed in future educational activities, or even on an individual HCP basis.

PredictCME

PredictCME is based on CHAID, which is often used in data mining. Unlike regression, CHAID can be used for both continuous and categorical data. Output is in the form of a classification (or decision) tree, which provides a visual representation of the interplay between predictor and response variables, as well as how the variable categories are broken down. Results from PredictCME will help guide needs assessments and ensure the appropriate topics, formats, questions, and audiences are targeted.

METHODS

Educational outcomes data were obtained from an educational activity on developing and implementing SAPs, which consisted of a faculty-led live and on-demand webcast, including a 60-minute panel discussion as well as a 30-minute Q&A. In addition, audio recordings of patient interviews were integrated into the content. HCP surveys assessing knowledge, confidence, and behavior were administered before, immediately following, and 3 months following the activity to establish baseline as well as any improvements as a result of the activity. A separate evaluation survey was also administered immediately following the activity, which provided demographics and other variables used in the model. An analysis using PredictCME was conducted on data from the pre-activity survey, which included a behavior question evaluating HCPs' promotion of active patient participation in shared decision-making when developing SAPs. Data from this behavior question were used as the response variable in the analysis, with demographics, knowledge, confidence, and evaluation data entered as predictors.

We decided we were interested in how often HCPs performed a behavior "at least 51% of the time," therefore data from the following behavior question were scored as follows. If participants selected "51% to 75%" or "76% to 100%," that was scored as a "1," and 0 otherwise.

1. In what percentage of your patients with epilepsy have you promoted their active participation using shared decision making to create a Seizure Action Plan?

a. 0%
b. 1% -25%
c. 26% -50%
d. 51% -75%
e. 76% -100%

Several predictor variables* were entered into the model:

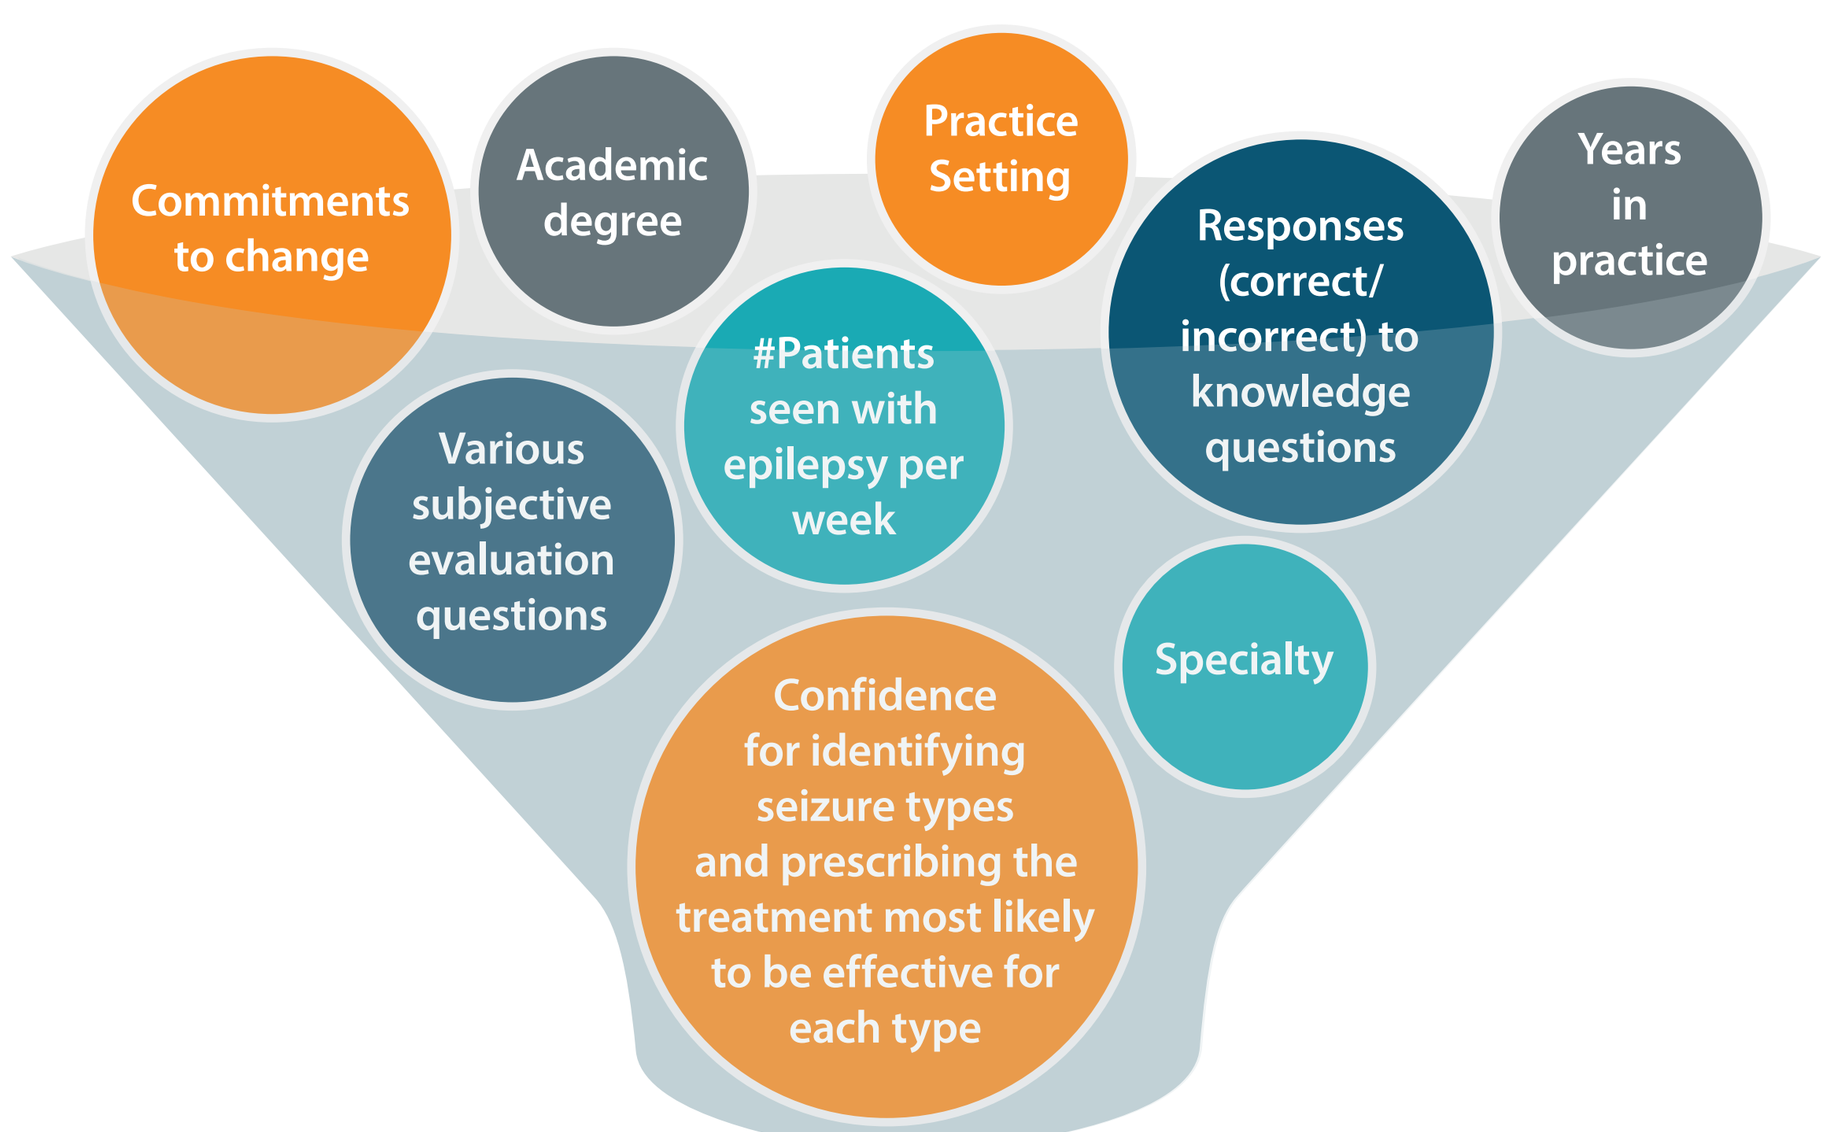
Commitments to change
Academic degree
Practice Setting
Responses (correct/ incorrect) to knowledge questions
Years in practice
Various subjective evaluation questions
#Patients seen with epilepsy per week
Specialty
Confidence for identifying seizure types and prescribing the treatment most likely to be effective for each type

* Predictor variables were selected based on expert assessments on which would most likely influence behavior and which variables would be of most interest. As CME Outfitters is the first provider to utilize this technique in medical education, there are currently no established algorithms or references guiding variable selection. We are in the process of developing such guidelines. In addition, although not available for this activity, data from questions related to practice barriers would be an important component of predictive models, which we will be incorporating in future PredictCME analyses. Finally, the CHAID algorithm prevents overfitting, so including multiple predictor variables (within reason) is not a significant concern.

RESULTS

Over 4,580 HCPs participated in the activity, with pre-survey data from 204 participants available for analysis. Figure 1 shows the breakdown of specialty, academic degrees and years in practice of the participants.

Figure 1. Demographic Distributions for Participants in an Educational Activity on Developing Seizure Action Plans for Patients with Epilepsy.

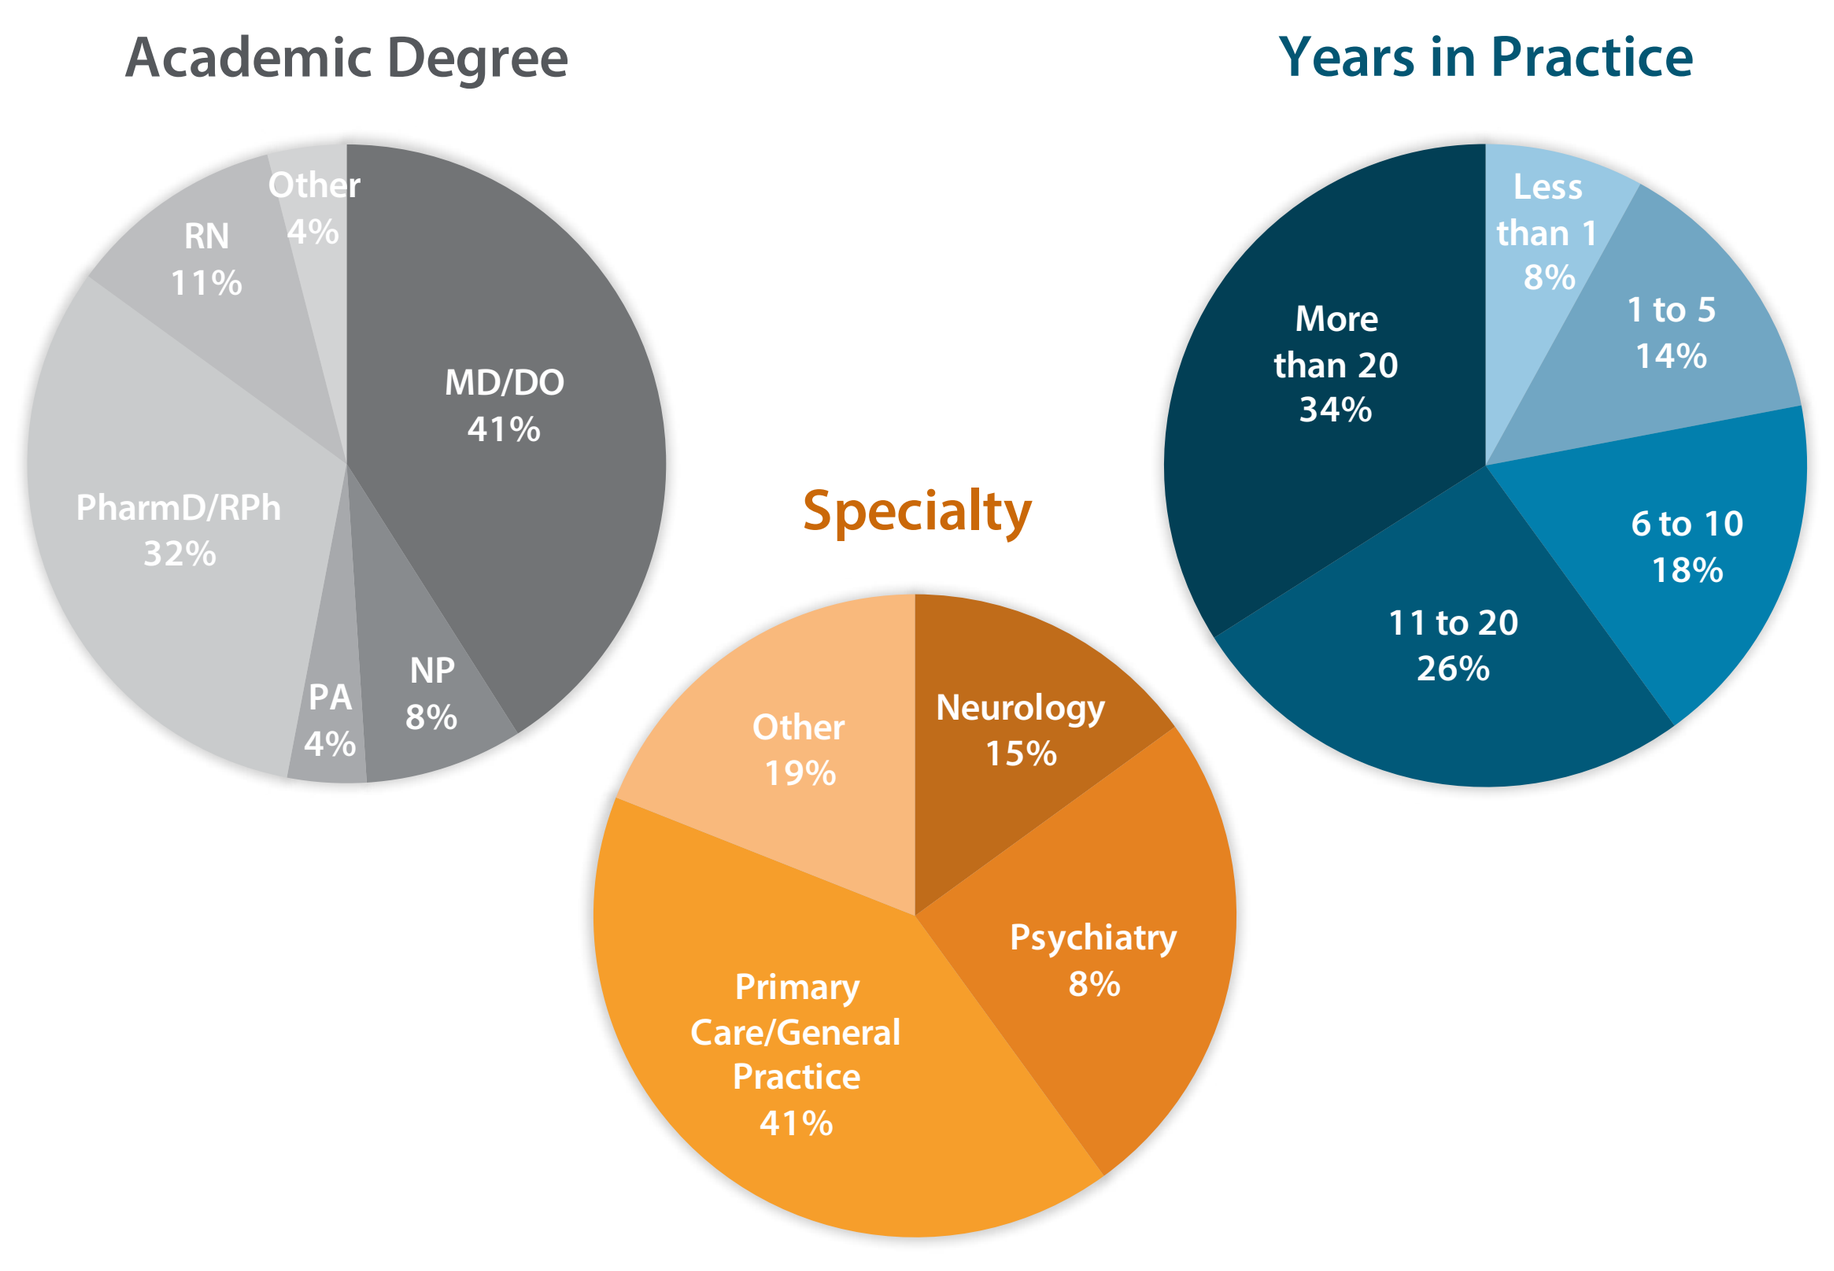
Academic Degree
MD/DO 41%
PharmD/RPh 32%
RN 11%
NP 8%
PA 4%
Other 4%
Specialty
Primary Care/General Practice 41%
Other 19%
Neurology 15%
Psychiatry 8%
Years in Practice
Less than 1 8%
1 to 5 14%
6 to 10 18%
11 to 20 26%
More than 20 34%

Figure 2 shows the PredictCME output for the pre-survey behavior response variable in tree format. All graphs in the output reflect percentages of participants who performed the behavior at least 51% of the time.

Figure 2. PredictCME Output for Pre-Survey Aggregate Behavior Score

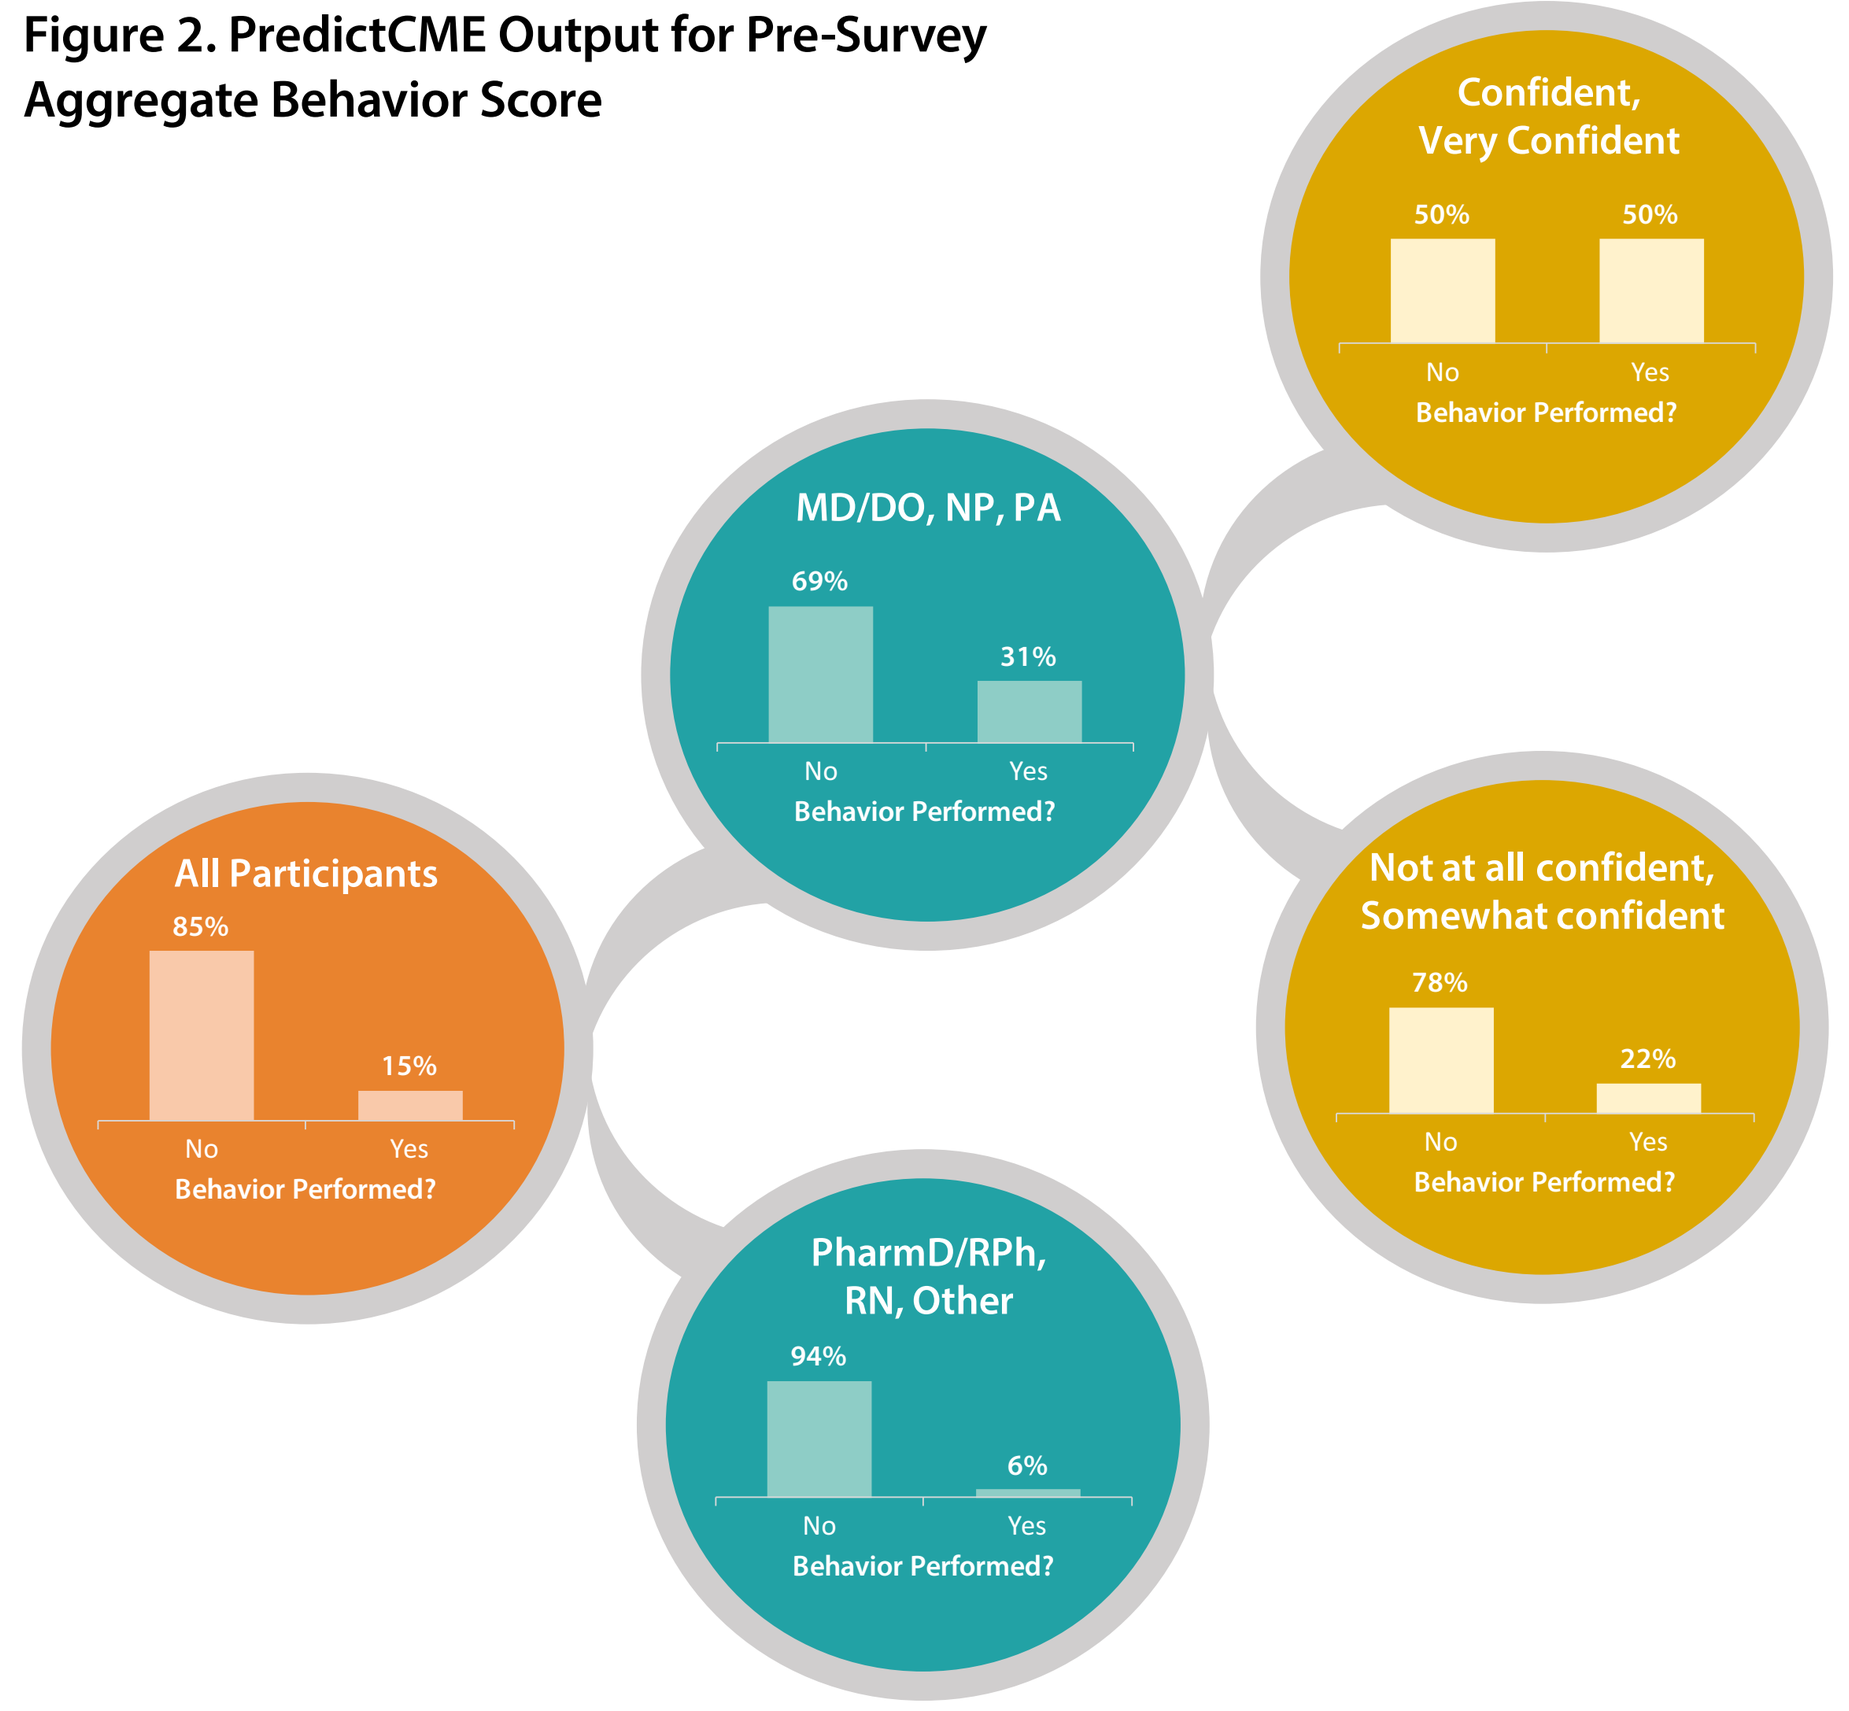
All Participants
85%
15%
No
Yes
Behavior Performed?
MD/DO, NP, PA
69%
31%
PharmD/RPh, RN, Other
94%
6%
Confident, Very Confident
50%
50%
Not at all confident, Somewhat confident
78%
22%

RESULTS (cont.)

Interpretation of the output would be as follows:

1. **Overall, *15% of participants* in the pre-survey indicated they promoted active participation of patients to create an SAP at least 51% of the time** (left-most graph)**.**

2. **The *primary*, or strongest, predictor of promoting active participation in patients to develop an SAP was *academic degree* ($\chi^2(1) = 23.47$, $p < .001$). MDs/DOs, NPs, and PAs were more likely than PharmD/RPhs, RNs, and HCPs with other degrees to perform the behavior at least 51% of the time** (31% vs. 6%, respectively, middle graphs).

3. **A *secondary* predictor was *confidence* in identifying seizure types and prescribing the most appropriate treatment ($\chi^2(1) = 5.67$, $p < .05$), with those who were more confident being more likely to perform the behavior than those who were less confident** (50% vs. 22%, respectively, right-most graphs)**. However, this predictor only applied to MDs/DOs, NPs, and PAs, thereby demonstrating an interaction in the model, as well as indicating that confidence did not influence the behaviors of PharmDs/RPhs, RNs, or HCPs with other degrees.**

CONCLUSIONS

Results from the PredictCME analysis are not surprising; NPs, MDs, DOs, and PAs have more opportunities to work directly with patients to develop SAPs and would therefore be expected to implement the behavior more often.

The secondary predictor of confidence is also not surprising, as prior studies have shown confidence to predict behavior.[2-6]

Taken together, these findings suggest that building HCP confidence is an important step toward encouraging best practices in developing SAPs with patients.

Future education can address HCP confidence, and perhaps sharing these findings with HCPs may also encourage them to reflect on ways they can increase utilization of SAPs in their practices.

These findings from the PredictCME analysis demonstrate the utility in using predictive modeling to better understand the influences of practice behavior. We prefer PredictCME to regression, as the procedure is more flexible, and the output is more intuitive and informative. It is our hope that other medical education providers will utilize predictive modeling, in its various forms, to help determine the factors that help or hinder the success of their educational activities, which in turn will help maximize the impact of future activities, and ultimately patient outcomes.

REFERENCE

1. Kass GV. An exploratory technique for investigating large quantities of categorical data. *Applied Statistics*. 1980;29(2):119-127.
2. Bandura A. Self-efficacy: toward a unifying theory of behavioral change. *Psychol Rev*. 1977;84:191-215.
3. Reiter J, Perez J, Tordoff S, Faler W. Novel Approaches to Predictive Modeling for Customizing Educational Activities. Poster presented at: SACME Annual Meeting; May 16-20, 2017; Scottsdale, AZ.
4. Reiter J, Perez J, Tordoff S, Faler W. Factors Influencing Best Practices in Treating Ulcerative Colitis: Results from a Predictive Modeling Analysis of Educational Outcomes Data. Poster presented at: World Congress of Gastroenterology @ACG2017; October 13-18, 2017; Orlando, FL.
5. Reiter J, Perez J, Tordoff S, Faler W. Factors Influencing Best Practices in Treating Multiple Sclerosis: Results from a Predictive Modeling Analysis. Poster accepted for presentation at: MSParis2017: 7th Joint ECTRIMS-ACTRIMS Meeting; October 25-28, 2017; Paris, France.
6. Reiter J, Perez J, Tordoff S, Faler W. Factors Influencing Best Practices in Treating IBD: Results from a Predictive Modeling Analysis of Educational Outcomes Data. Poster presented at: Advances in Inflammatory Bowel Disease (AIBD) Annual Meeting; November 9-11, 2017; Orlando, FL.

FUNDING

The educational activity described in this presentation was supported by an educational grant from Upsher-Smith Laboratories, Inc.

CME Outfitters
CONTINUING MEDICAL EDUCATION

Scan the QR code on the right to view a pdf of this poster online.

© CME Outfitters, LLC, 2017. All rights reserved.